Co-Administration of the Oral S1P-Lyase Inhibitor LX2931 (LX3305) with Methotrexate was Well Tolerated Over 14 Days in Patients with Stable Rheumatoid Arthritis

Roy Fleischmann[1], Kenny S. Frazier[2], Joel Freiman[2], Barbara Brooks[2], Michael S. Donoviel[2], Tamas Oravecz[2], David Augeri[3], William E. Heydorn[3], Michael Kelly[2], and Philip Brown[2]
[1]Metroplex Clinical Research Center, Dallas, TX, United States 75231, [2]Lexicon Pharmaceuticals, Inc., The Woodlands, TX, United States 77381, and [3]Lexicon Pharmaceuticals, Inc., Princeton, NJ, United States 08540

426

INTRODUCTION

Sphingosine 1-phosphate (S1P) is a biologically abundant lipid mediator that plays an essential role in cell signaling pathways, particularly in the immune system where changes in local S1P concentration and gradient can alter inflammatory cell responses,[1,2] affect the barrier function of endothelial cells,[(3-5)] and modify lymphocyte migration patterns.[(6-10)] The concentration gradient of S1P between lymphoid tissues and the peripheral blood is tightly regulated by both metabolic and catabolic enzymes. Sphingosine-1-phosphate lyase 1 (S1PL or SGPL1) is the main enzyme responsible for the degradation of S1P (**Figure 1**). Mice with reduced S1PL activity have significantly reduced circulating lymphocytes as a consequence of increased S1P content in lymphoid tissues.[11]

LX2931, also known as LX3305, is an orally-bioavailable small molecule inhibitor of S1PL (**Figure 1**) being developed by Lexicon Pharmaceuticals, Inc., as a potential therapeutic compound for the treatment of autoimmune and inflammatory disorders, such as rheumatoid arthritis (RA). In preclinical studies, LX2931 demonstrated potent anti-inflammatory activity in rodent models of arthritis, inflammation and transplantation. In Phase 1 studies, LX2931 was well tolerated at all dose levels tested and produced a dose-dependent reduction in circulating lymphocytes. **In the current study, discussed here, co-administration of MTX with LX2931 was well tolerated over 14 days of dosing in patients with stable rheumatoid arthritis (RA). These results further support that inhibition of S1PL with a small molecule therapy, such as LX2931, may represent a new mechanism for immune modulation with the potential to address inflammatory diseases such as RA as well as other autoimmune and inflammatory disorders.**

Figure 1. LX2931 regulates lymphocytes by targeting S1P lyase, the enzyme that degrades S1P.

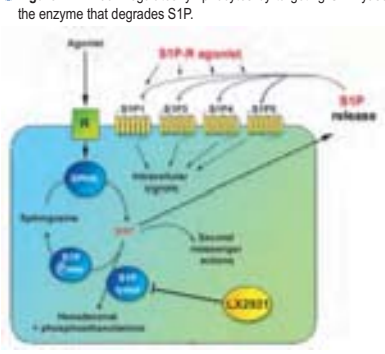
Agonist
S1P-R agonist
S1P1
S1P3
S1P4
S1P5
S1P release
LX2931

PRECLINICAL RESULTS

- A novel drug target (S1PL) was discovered through Lexicon's high-throughput mouse knockout phenotyping program (Genome 5000™).
- Genetic inhibition of S1PL through the use of knockout mouse technology demonstrates significant reduction in circulating lymphocytes.[11]
- LX2931 recapitulates the lymphopenic phenotype observed in knockout mice in a dose-dependent manner in rodents and primates (**Figure 2**).
- Physiologic response to LX2931 treatment reveals a high tissue specificity as demonstrated by significant increases of S1P in the thymus, spleen, and jejunum compared to other tissues (**Figure 3**).
- LX2931 treatment delays allograft rejection and ameliorates the development of inflammation in multiple animal models of disease, including rodent models of rheumatoid arthritis (**Figure 4**).
- LX2931 shows therapeutic potency at doses where lymphopenia is not observed (**Figure 5**).
- LX2931 potentiates the therapeutic effect of Methotrexate (MTX) in a rat adjuvant arthritis model (**Figure 6**).
- Anti-inflammatory effects of LX2931 are not due to a generalized immunosuppression (**Figure 7**).

Figure 2. LX2931 mechanism of action is conserved from mice to primates.

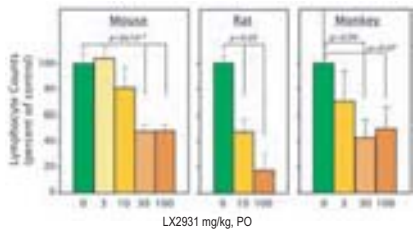
Mouse
Rat
Monkey
Lymphocyte Counts (percent of control)

LX2931 mg/kg, PO

LX2931 was administered as a single dose 18 hrs (rodent) or 72 hrs (monkey) prior to blood draw; data are presented as mean +/- SEM. (n=4-10 animals per cohort)

Figure 3. LX2931 elevates S1P levels primarily in the lymphoid system.

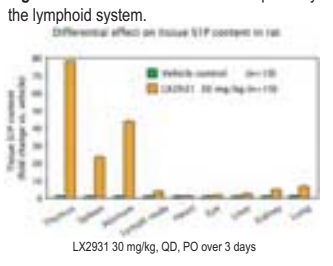
Differential effect on tissue S1P content in rat

LX2931 30 mg/kg, QD, PO over 3 days

Tissues were evaluated 18 hrs after last dose. Data were pooled from two experiments using 4-8 animals in each group. Data are presented as mean +/- SEM.

PRECLINICAL RESULTS (continued)

- **Figure 4.** LX2931 inhibits inflammatory response in a prophylactic model of arthritis.

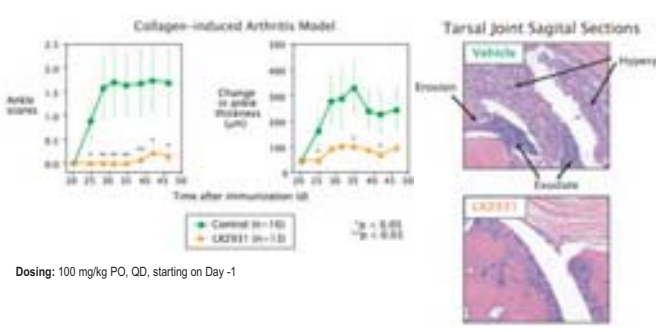
Collagen-induced Arthritis Model
Tarsal Joint Sagittal Sections
Vehicle
LX2931
Erosion
Hyperplasia
Exudate
Time after immunization (d)

Dosing: 100 mg/kg PO, QD, starting on Day -1

- **Figure 5.** LX2931 inhibits inflammatory response in a treatment model of arthritis in mice.

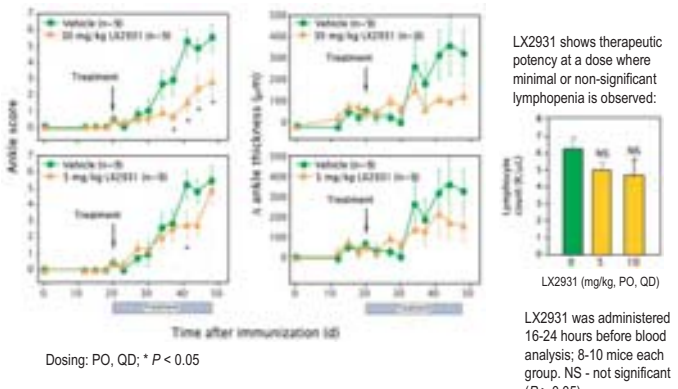
Treatment
Ankle score
Time after immunization (d)

Dosing: PO, QD; * $P < 0.05$

LX2931 shows therapeutic potency at a dose where minimal or non-significant lymphopenia is observed:

LX2931 (mg/kg, PO, QD)

LX2931 was administered 16-24 hours before blood analysis; 8-10 mice each group. NS - not significant ($P > 0.05$).

- **Figure 6.** LX2931 potentiates therapeutic effect of methotrexate in animal model of arthritis.

Rat Adjuvant-Induced Arthritis Model

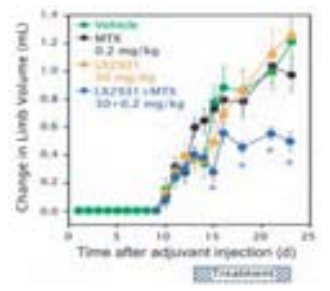
Vehicle
MTX 0.2 mg/kg
LX2931 30 mg/kg
LX2931+MTX 30+0.2 mg/kg
Change in Limb Volume (mL)
Time after adjuvant injection (d)
Treatment

Treatments were PO, QD. Data are presented as mean +/- SEM (n = 9). * $P < 0.05$

- **Figure 7.** LX2931 does not cause general immunosuppression even at a dose that produces altered lymphocyte trafficking.

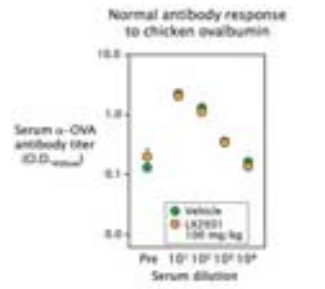
Normal antibody response to chicken ovalbumin
Serum α-OVA antibody titer
Vehicle
LX2931 100 mg/kg
Serum dilution

LX2931 treated mice showed normal *in vivo* antibody responses when subjected to T-cell dependent systemic challange with chicken ovalbumin. Mice were dosed PO, QD starting one day prior to administration of ovalbumin. Anti-ovalbumin IgG1 antibody titers in serum were measured on Day 14, 18 hours after the last dose. Data are presented as mean +/- SEM. Pre, pre-immune sera diluted 1:10.

PHASE 1 STUDY RESULTS

- Two single-dose and two multiple-dose, placebo-controlled trials have been completed in healthy volunteers.
 - Liquid and solid oral dose formulations behave similarly
 - Dose ranges tested from 10 mg to 250 mg
 - 108 active treatment participants; 36 placebo participants
- Pharmacokinetic analyses support a once-daily dosing regimen, with steady-state levels achieved by Day 4 of dosing.
- Dose-dependent reduction in absolute lymphocyte counts (ALC) consistently observed throughout all studies, confirming preclinical results (**Figure 8**).
- Rapid onset and offset of pharmacologic activity: nadirs in ALC, as well as T- and B-cell subpopulations, observed 24 hours after single dose; return to baseline levels within 48-72 hours following treatment discontinuation
- Maximal lymphopenic effect observed by the 150 mg dose level (**Figure 9**), which was well tolerated over 7 days of dosing.
- Good safety profile, with most commonly reported treatment emergent adverse events (TEAEs) assessed as mild in intensity; two subjects in the single-dose Phase 1 studies, one at 180 mg QD and one at 250 mg (125 mg BID), developed an acute and transient right upper quadrant pain resembling biliary colic, accompanied by elevations in ALT and AST without concomitant elevation in bilirubin.
- No clinically significant changes in vital signs (including heart rate) or ECG's have been observed.

Figure 8. LX2931 produces a dose-dependent reduction in lymphocyte counts in normal subjects.

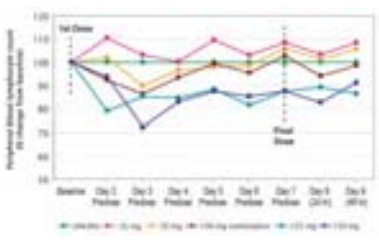

Figure 9. LX2931 produces a maximal lymphopenic effect by the 150 mg dose level in normal subjects.

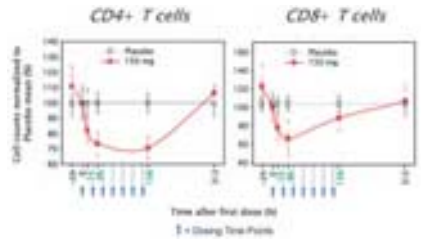
CD4+ T cells
CD8+ T cells

Drug-Drug Interaction (DDI) STUDY RESULTS

- Conducted at a single site (Dr. Roy M. Fleischmann, Metroplex Clinical Research Center) in Dallas, Texas.
- Randomized, double-blind, placebo-controlled study that evaluated a single daily dose of LX2931 (120 mg), co-administered with MTX, over a 14-day period in patients with stable rheumatoid arthritis (**Table 1**; **Figure 10**).
- Study treatment was safe and well tolerated, with no evidence of enhanced MTX toxicity in the presence of LX2931.
- The majority of TEAEs were mild in intensity, with GI disorders being the most commonly cited body system (**Table 2**); no serious adverse events were reported.
- No significant trends were apparent for changes in laboratory values, vital signs, physical findings, or ECGs.
- A modest reduction in lymphocyte counts was observed at the conclusion of the treatment period when compared to baseline.
- No clinically relevant changes were observed for the kinetic parameters of LX2931, MTX, or its metabolite 7-OH-MTX (**Figure 11**).
- No changes were observed in the amount of MTX or 7-OH-MTX excreted in urine, indicating that multiple-dose administration of LX2931 did not have an effect on renal clearance.
- No accumulation of LX2931 was detected following repeated dosing.

Figure 10. Design for LX2931 + MTX drug-drug interaction study.

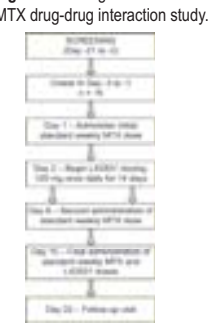

Table 1. DDI Patient Demographics

	LX2931 (n=12)	Placebo (n=3)	All Subjects (n=15)
Age (years)			
Mean (SD)	59.8 (10.25)	58.7 (14.74)	59.5 (10.67)
Median (min-max)	63.0 (35-70)	64.0 (42-70)	63.0 (35-70)
Gender (n,%)			
Female	12 (100.0%)	3 (100.0%)	15 (100.0%)
Race (n,%)			
Caucasian (White)	9 (75.0%)	3 (100.0%)	12 (80.0%)
Black or African American	3 (25.0%)	0 (0.0%)	3 (20.0%)
Hispanic or Latino Ethnicity (n,%)			
Yes	2 (16.7%)	0 (0.0%)	2 (13.3%)
No	10 (83.3%)	3 (100.0%)	13 (86.7%)
BMI at Screening (kg/m^2)			
Mean (SD)	31.32 (7.779)	26.53 (7.006)	30.36 (7.647)
Median (min-max)	29.65 (24.1-48.7)	26.20 (19.7-33.7)	28.90 (19.7-48.7)
DAS28-CRP Score at Screening			
Mean (SD)	2.647 (0.7451)	2.415 (1.4113)	2.601 (0.8544)
Median (min-max)	2.615 (1.48-3.86)	2.031 (1.24-3.98)	2.590 (1.24-3.98)

Figure 11. Pharmacokinetic profile for LX2931+ MTX DDI Study

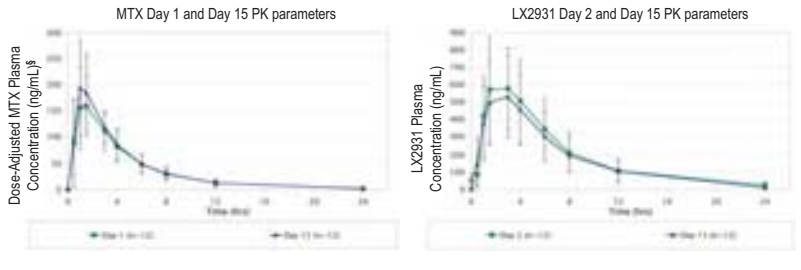
MTX Day 1 and Day 15 PK parameters
LX2931 Day 2 and Day 15 PK parameters
Dose-Adjusted MTX Plasma Concentration (ng/mL)§
LX2931 Plasma Concentration (ng/mL)

§ MTX dose-adjusted parameter (adjusted to 7.5 mg)

Table 2. DDI Treatment Emergent Adverse Event Summary

Body System Preferred Term	LX2931 (n=12) Subjects, (%)	Placebo (n=3) Subjects, (%)	Overall (n=15) Subjects, (%)
Gastrointestinal Disorders	5 (41.7)	0 (0.0)	5 (33.3)
General Disorders and Site Administration Conditions	4 (33.3)	0 (0.0)	4 (26.7)
Infections and Infestations	3 (25.0)	1 (33.3)	4 (26.7)
Nervous System Disorders	3 (25.0)	1 (33.3)	4 (26.7)
Musculoskeletal and Connective Tissue Disorders	2 (16.7)	1 (33.3)	3 (20.0)
Eye Disorders	1 (8.3)	0 (0.0)	1 (6.7)
Metabolism and Nutrition Disorders	0 (0.0)	1 (33.3)	1 (6.7)
Respiratory, Thoracic, and Mediastinal Disorders	2 (16.7)	0 (0.0)	2 (13.3)

CONCLUSIONS

- **LX2931 (a.k.a., LX3305) is an orally-bioavailable small molecule inhibitor of S1PL being developed as a potential therapeutic compound for the treatment of autoimmune and inflammatory disorders, such as RA.**
- **In preclinical studies, LX2931 demonstrated potent anti-inflammatory activity in rodent models of arthritis, inflammation and transplantation.**
- **Co-administration of MTX with LX2931 was well tolerated over 14 days of dosing in patients with stable RA.**
- **The most commonly occurring adverse events included abdominal pain, nausea, and headache. There were no clinically significant trends observed in laboratory values, vital signs, or electrocardiograms.**
- **No clinically significant changes in either MTX or LX2931 kinetics were observed.**
- **Inhibition of S1PL with a small molecule therapy, such as LX2931, represents a new mechanism for immune modulation with the potential to address inflammatory diseases such as RA.**
- **A Phase 2 study to determine the safety and efficacy of daily orally administered LX2931 in subjects with active RA on stable MTX therapy is currently ongoing.**

REFERENCES:
1. Liao JJ, Huang MC, Goetzl EJ. Cutting edge: Alternative signaling of Th17 cell development by sphingosine 1-phosphate. J Immunol. 2007;178:5425-5428.
2. Pettus BJ, Chalfant CE, Hannun YA. Sphingolipids in inflammation: roles and implications. Curr Mol Med. 2004;4:405-418.
3. McVerry BJ, Garcia JG. In vitro and in vivo modulation of vascular barrier integrity by sphingosine 1-phosphate: mechanistic insights. Cell Signal. 2005;17:131-139.
4. Sanna MG, Wang SK, Gonzalez-Cabrera PJ, Don A, Marsolais D, et al. Enhancement of capillary leakage and restoration of lymphocyte egress by a chiral S1P1 antagonist in vivo. Nat Chem Biol. 2006;2:434-441.
5. Wei SH, Rosen H, Matheu MP, Sanna MG, Wang SK, et al. Sphingosine 1-phosphate type 1 receptor agonism inhibits transendothelial migration of medullary T cells to lymphatic sinuses. Nat Immunol.
6. Cyster JG. Chemokines, sphingosine-1-phosphate, and cell migration in secondary lymphoid organs. Annu Rev Immunol. 2005;23:127-159.
7. Rosen H, Goetzl EJ. Sphingosine 1-phosphate and its receptors: an autocrine and paracrine network. Nat Rev Immunol. 2005;5:560-570.
8. Schwab SR, Cyster JG. Finding a way out: lymphocyte egress from lymphoid organs. Nat Immunol. 2007;8:1295-1301.
9. Schwab SR, Pereira JP, Matloubian M, Xu Y, Huang Y, et al. Lymphocyte sequestration through S1P lyase inhibition and disruption of S1P gradients. Science. 2005;309:1735-1739.
10. Shiow LR, Rosen DB, Brdickova N, Xu Y, An J, et al. CD69 acts downstream of interferon-alpha/beta to inhibit S1P1 and lymphocyte egress from lymphoid organs. Nature. 2006;440:540-544.
11. Vogel P, Donoviel MS, Read R, Hansen GM, Hazlewood J, et al. Incomplete inhibition of sphingosine 1-phosphate lyase modulates immune system function yet prevents early lethality and non-lymphoid lesions. PLoS One. 2009;4(1):e4112.

DISCLOSURE: The authors[(2,3)] of this poster are employees of, and have received stock options from, Lexicon Pharmaceuticals, Inc.
The presenting author[1] of this poster has received compensation from Lexicon Pharmaceuticals, Inc. both as a consultant and clinical investigator.

Presented at the 2009 American College of Rheumatology Meeting - Philadelphia, PA - October 18th 2009